EVALUATION OF A MINIMALLY INVASIVE DATA LOGGER FOR COLLECTION OF HEART RATE AND BODY TEMPERATURE IN CYNOMOLGUS MONKEYS

Jason Cordes, Patrick Malloy, Jill Steidl-Nichols – Global Safety Pharmacology
Pfizer Worldwide Research & Development, Groton Laboratories, Groton, CT 06340

3044

ABSTRACT

Heart rate (HR) and body temperature (BT) can be important physiological parameters to monitor on toxicology studies. Traditionally, surface lead ECGs and rectal thermometers are used to collect these data, but only provide a snapshot of data at any one time. Telemetry add-ons can also be utilized to capture these data, but this level of investment may not be warranted in all cases. In this study, Star Oddi (SO) DST milli-HRT data loggers were implanted in 4 telemetry-instrumented cynomolgus monkeys and evaluated for use as a potential toxicology add-on technology for measuring HR and BT in monkeys. All surgeries (implantations) were well tolerated with minimal recovery time needed. The animals were administered vehicle or 2 mg/kg diazepam in a crossover design. Cardiovascular (CV) parameters were recorded for 24 hours postdose (HPD) by both telemetry and SO loggers, and rectal temperatures were collected predose and 4 HPD. Upon study completion, the data loggers were explanted from the animals to retrieve the data. The SO loggers produced high quality data, as 77% of the data was deemed reliable based upon visual review. Both recording technologies detected statistically significant decreases in HR (-19 to -28 bpm) and BT (-0.4 to -0.5°C) from 0.5-3.5 HPD. No statistically significant differences were observed between the SO and telemetry data. Decreases in BT were similar between rectal, telemetry and SO collection methods. The results of this study suggest that the SO DST milli-HRT logger is a viable minimally invasive add-on technology for HR and BT measurement.

METHODS

All procedures performed on these animals were in accordance with regulations and established guidelines and were reviewed and approved by Pfizer's Institutional Animal Care and Use Committee

The Star Oddi DST milli-HRT (Fig. 1) is a small data logger that can be implanted subcutaneously or intramuscularly to measure HR and BT. The SO data logger records data at configurable intervals, and stores the data internally. The SO logger must be explanted to retrieve the data.

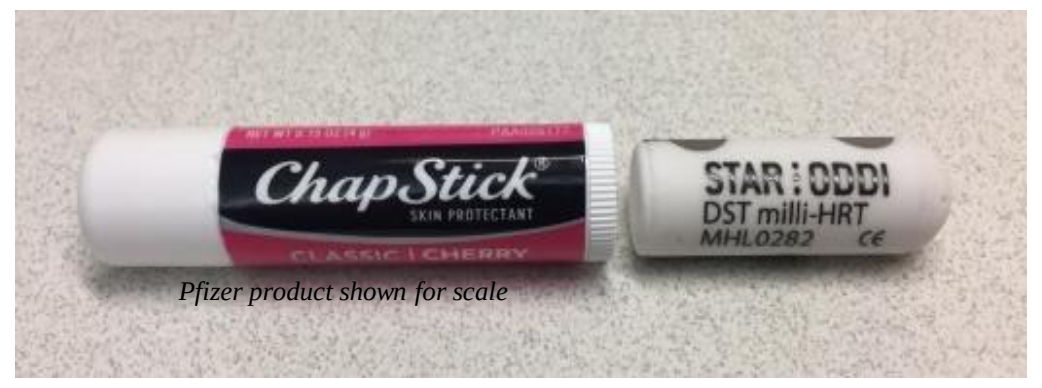
ChapStick
SKIN PROTECTANT
CLASSIC | CHERRY
STAR:ODDI
DST milli-HRT
MHL0282
Pfizer product shown for scale

Figure 1: SO DST milli-HRT Data Logger

Four male cynomolgus monkeys (7.7-8.6 kg), previously implanted with traditional telemetry devices (Data Sciences International), were chosen for the study. The SO logger was implanted subcutaneously in the lower right abdominal area in animals 2 and 4 and intramuscularly between the internal and external abdominal oblique in animals 1 and 3. The monkeys were then allowed 8 days to recover before dose administration.

Diazepam was used as the test article in order to induce a measurable HR and BT decrease. The monkeys were orally dosed with both vehicle and 2 mg/kg of diazepam in a single dose crossover study design (Table 1). Animals were allowed 5 days between doses to allow sufficient clearance of compound.

Animal Number	Treatment 1 (CV) mg/kg	Treatment 2 (CV) mg/kg
1	0	2
2	0	2
3	2	0
4	2	0

Table 1: Dosing Design. CV = Cardiovascular; Animals were dosed at approximately 9:00AM. Vehicle was 0.1% methylcellulose.

METHODS (cont'd)

The traditional telemetry system recorded data continuously from approximately 1 hour predose to 24 hours postdose (HPD). The SO data logger collected data at specific intervals described in Table 2. ECG electrodes on the SO logger surface recorded the ECG (from which HR was derived) for a burst of 3 seconds at each recording interval. HR data were rated by the SO system on a Quality Index (QI) from 0 (best) to 3 (worst).

Time	Data Interval
-1 to 4 HPD	1 minute
4 to 24 HPD	15 minutes
24 HPD to next dose	1 hour

Table 2: SO Recording Intervals

Telemetry data were collected using Ponemah (DSI) and analyzed using ecgAuto (emka Technologies). SO logger data were downloaded using Mercury software. Both data sets were then further analyzed with Microsoft Excel, and statistics (ANOVA) were calculated using SAS 9.4.

Rectal body temperatures were obtained predose and at 4 HPD.

RESULTS

All surgeries were successful with minimal recovery time. Minimal initial signs of skin erosion were seen in subcutaneous implantation. All loggers were explanted and data downloaded without incident.

SO loggers successfully recorded the ECG and BT temperature data. Based upon visualization/analysis of ECG and HR data, only data with a QI of 0 and 1 were included in the analysis (Fig. 2).

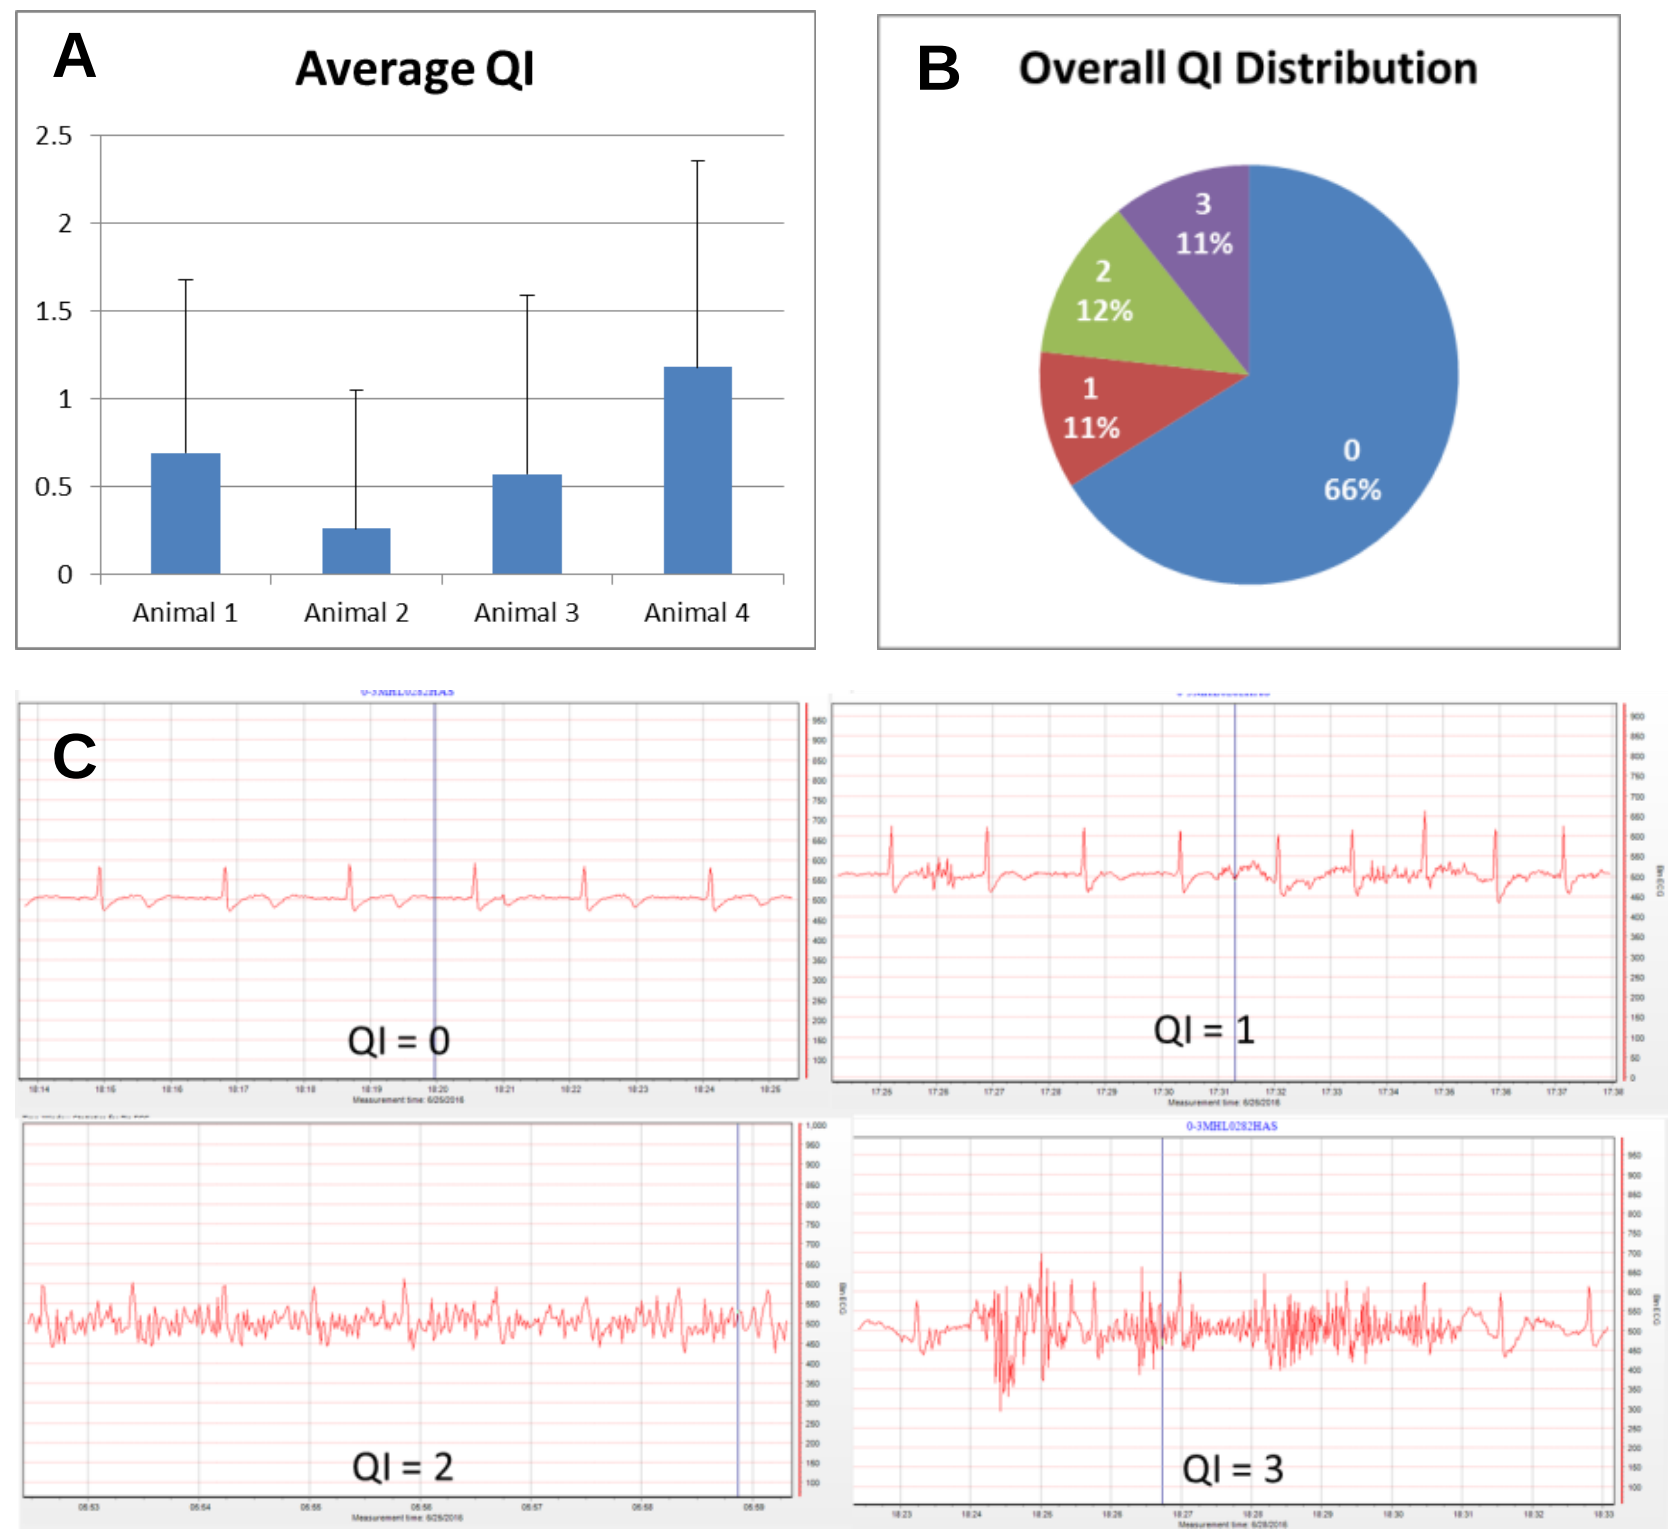
A
Average QI
2.5
2
1.5
1
0.5
0
Animal 1
Animal 2
Animal 3
Animal 4
B
Overall QI Distribution
3
11%
2
12%
1
11%
0
66%
C
QI = 0
QI = 1
QI = 2
QI = 3

Figure 2: Quality Index. **A:** Average QI for each animal. **B:** Distribution of QI for entire study. **C:** Sample ECG waveforms for each QI level.

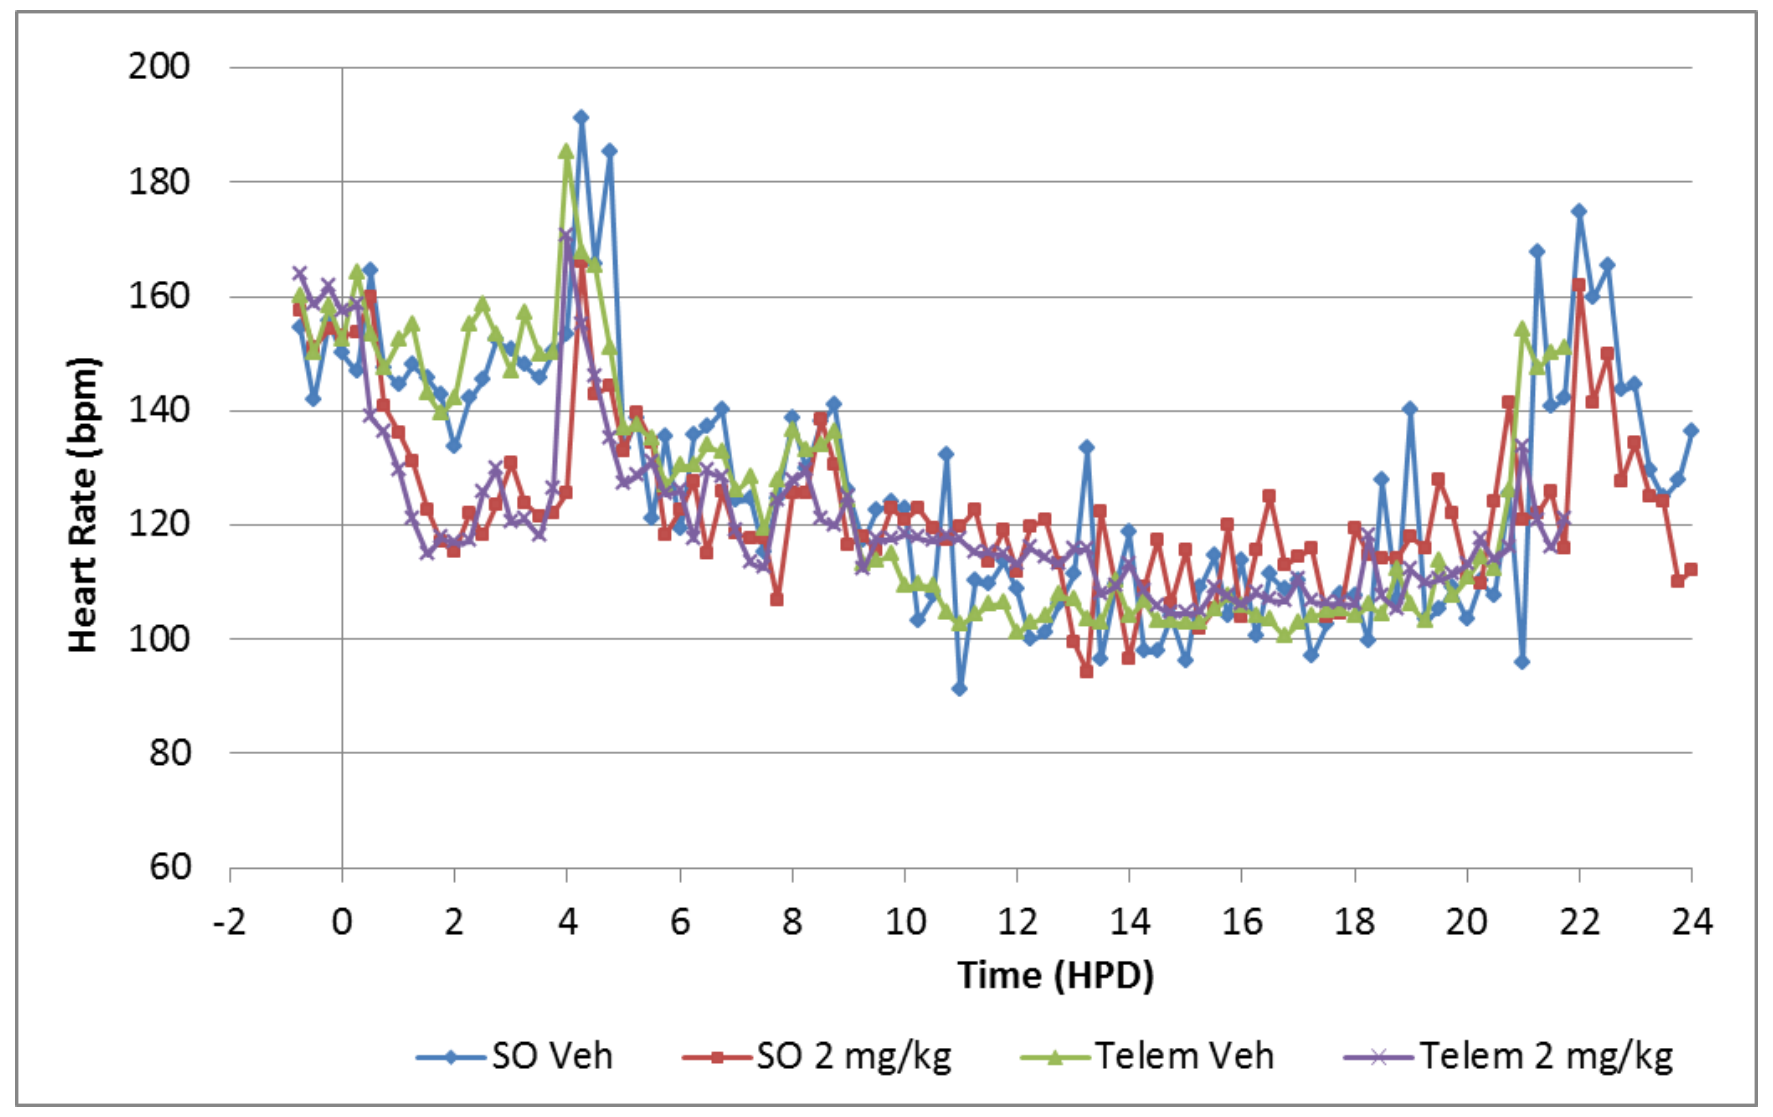
Heart Rate (bpm)
200
180
160
140
120
100
80
60
-2
0
2
4
6
8
10
12
14
16
18
20
22
24
Time (HPD)
SO Veh
SO 2 mg/kg
Telem Veh
Telem 2 mg/kg

Figure 3: Heart Rate Data. Each symbol represents the 15-min average of 4 animals. Dose administration is at 0 HPD. bpm = beats per minute; Veh = vehicle.

HR data were similar between SO and telemetry data, especially during the 1 min recording period (Predose up to 4 HPD). The 15 minute recording period was prone to spikes and dropout due to QI filtering (Figure 3). Statistical significance was achieved in first time bin for both data sets [-18.9, -27.7 bpm, SO and telemetry, respectively]. Although the mean difference with the SO logger was smaller, no statistically significant difference was found between SO and telemetry data (Table 3).

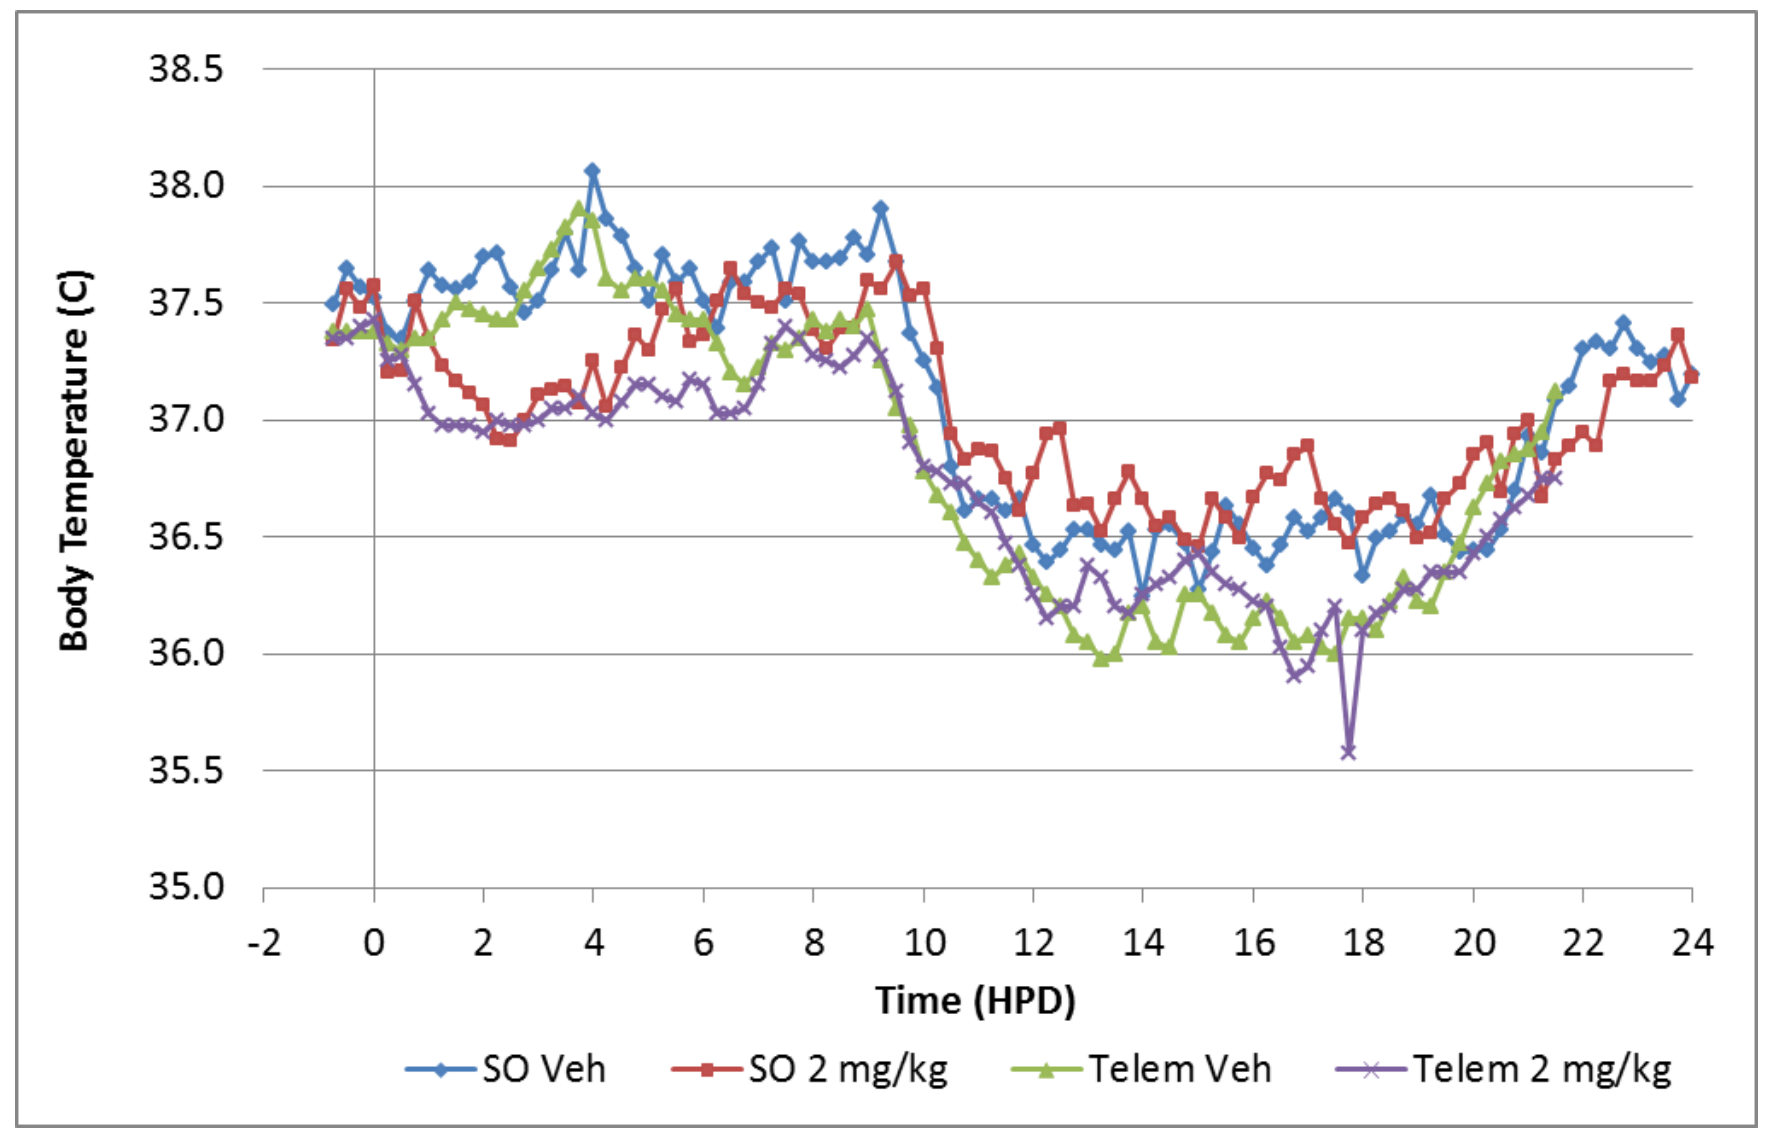
Body Temperature (C)
38.5
38.0
37.5
37.0
36.5
36.0
35.5
35.0
-2
0
2
4
6
8
10
12
14
16
18
20
22
24
Time (HPD)
SO Veh
SO 2 mg/kg
Telem Veh
Telem 2 mg/kg

Figure 4: Body Temperature Data. Each symbol represents the 15-min average of 4 animals. Dose administration is at 0 HPD. Veh = vehicle

Decreased body temperature from 0.5 – 3.5 HPD was observed in both SO [-0.4 °C] and telemetry [-0.5 °C] data, as expected due to diazepam's known effects (Figure 4, Table 3). No statistically significant difference was found between SO and telemetry data. Rectal temperature was consistently higher, but all 3 methods detected similar changes in BT (-0.8 to -0.9 °C at 4 HPD vs vehicle; Figure 5).

Additionally, the telemetry system detected statistically significant differences in QT and RR intervals, consistent with HR decreases. A decrease in activity was detected, as expected based on known effects of diazepam. These parameters are not recorded by the SO system (Table 3).

Significant Differences- 0.5 to 3.5 HPD	SO Logger	Telemetry
Heart Rate (bpm)	-18.9	-27.7
Temperature (°C)	-0.4	-0.5
Activity (arbitrary units)	N/A	-10.8
SO vs. Telemetry: HR	-	
SO vs. Telemetry: BT	-	

Table 3: Selection of statistically significant differences between vehicle and treatment data. bpm = beats per minute.

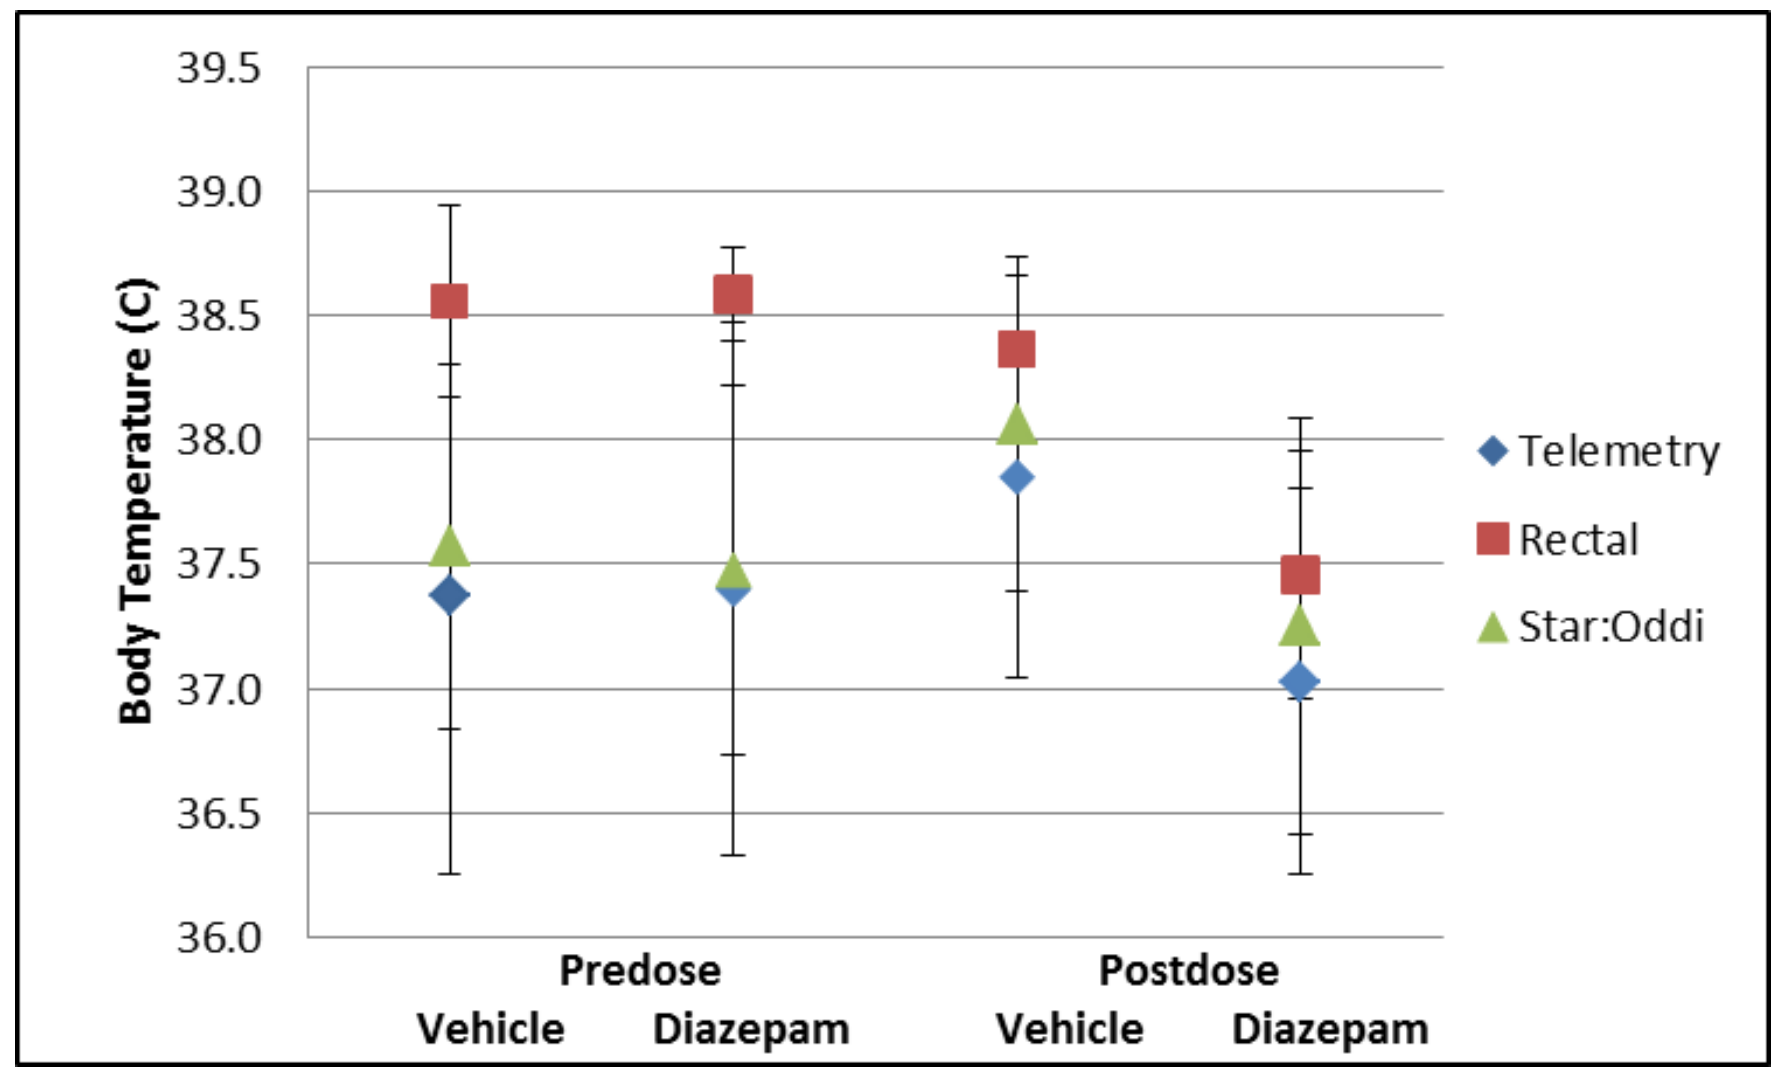
Body Temperature (C)
39.5
39.0
38.5
38.0
37.5
37.0
36.5
36.0
Predose
Postdose
Vehicle
Diazepam
Vehicle
Diazepam
Telemetry
Rectal
Star:Oddi

Figure 5: Body temperature changes from predose to 4 HPD, as measured by telemetry, SO, and rectal thermometers.

CONCLUSIONS

- The SO logger was found to be minimally invasive and well tolerated by the monkeys.
- SO detected decreases in HR and BT that were similar to those recorded by the telemetry system and rectal thermometers.
- Benefits:
 - Minimally invasive, low cost
 - No ECG analysis required for HR measurement
- Limitations:
 - Implant must be programmed prior to implantation, which limits flexibility
 - No real time data visualization; explanation required
- Recommendations:
 - Only HR data with a QI of 0 or 1 should be accepted.
 - Minimize data collection intervals for greatest accuracy.
 - Intramuscular placement is preferred over subcutaneous placement due to skin erosion.
- The SO logger is a potentially useful add-on technology when HR and BT measurements are desired.

ACKNOWLEDGMENTS

Safety Pharmacology: Peter Harris, David Ackley (sponsor)
Statistical analysis: Dingzhou Li
Comparative Medicine: Jan Bernal, Steven Kreuser, Marlo Volberg, Evan Pagano, Amy Martunas, Sue Rubino, Larry Matney, Chris Olszewski
Star Oddi for providing technical support.